The Farnesoid X Receptor (FXR) Agonist EDP-305 Inhibits Fibrosis Progression in a Rat Model of Nonalcoholic Steatohepatitis Cirrhosis

Sarani Ghoshal[1], Gunisha Arora[1], Ricard Masia[2], Diego S. Ferriera[3], Mozhdeh Sojoodi[1], Yang Li[4], Guogiang Wang[4], Yat Sun Or[4], Li-Juan Jiang[4], Kenneth K. Tanabe[1], Bryan C. Fuchs[1]

[1]Division of Surgical Oncology, [2]Department of Pathology, [3]Martinos Center for Biomedical Imaging, Massachusetts General Hospital and Harvard Medical School, Boston, MA, United States
[4]Enanta Pharmaceuticals, Inc., Watertown, MA, USA

Background: Farnesoid X Receptor

Farnesoid X receptor (FXR) is a nuclear receptor that has emerged as a key regulator in the maintenance of bile acid homeostasis.

FXR agonists are currently under clinical investigation for the management of various liver diseases such as primary biliary cholangitis (PBC) and nonalcoholic steatohepatitis (NASH).

However, most preclinical studies have examined the effects of FXR agonists in mouse models where fibrosis progression to cirrhosis is challenging.

Here, we investigate the anti-fibrotic effects of the FXR agonist EDP-305 in a rat model of NASH cirrhosis.

CDAHFD Fed Rats Develop Bridging Fibrosis and Cirrhosis Reminiscent of Late-Stage F3-4 Human Disease

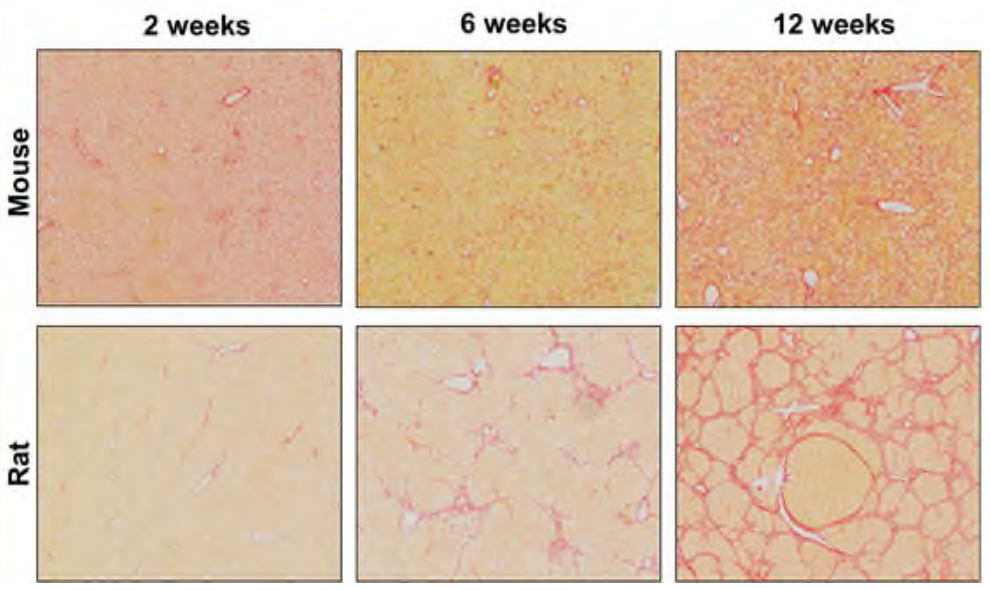
2 weeks
6 weeks
12 weeks
Mouse
Rat

When fed to mice, the L-amino acid-defined, high fat diet with 60 kcal% fat and 0.1% methionine (CDAHFD) induces a severe fibrotic response. However, the collagen deposition patterns in a “chicken-wire” format similar to other mouse models and reminiscent of early human disease. By comparison, rats fed the CDAHFD developed bridging fibrosis by 6 weeks that progressed to nodular cirrhosis by 12 weeks, reminiscent of late-stage human disease.

CDAHFD-Induced Rat Model of NASH Cirrhosis with EDP-305 Treatment

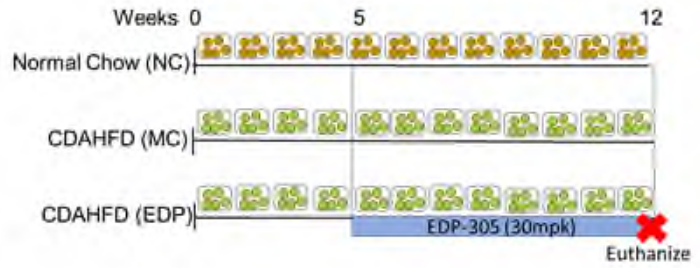
Weeks 0 5 12
Normal Chow (NC)
CDAHFD (MC)
CDAHFD (EDP)
EDP-305 (30mpk)
Euthanize

Male Wistar rats (175-200 g) were fed either normal chow (NC) or CDAHFD for 12 weeks. Animals were randomized to receive either EDP-305 30mg/kg or vehicle control (0.5% methylcellulose (MC)) at the start of week 5 (n = 8 per group).

EDP-305 Treatment was Well Tolerated in the CDAHFD Rat Model

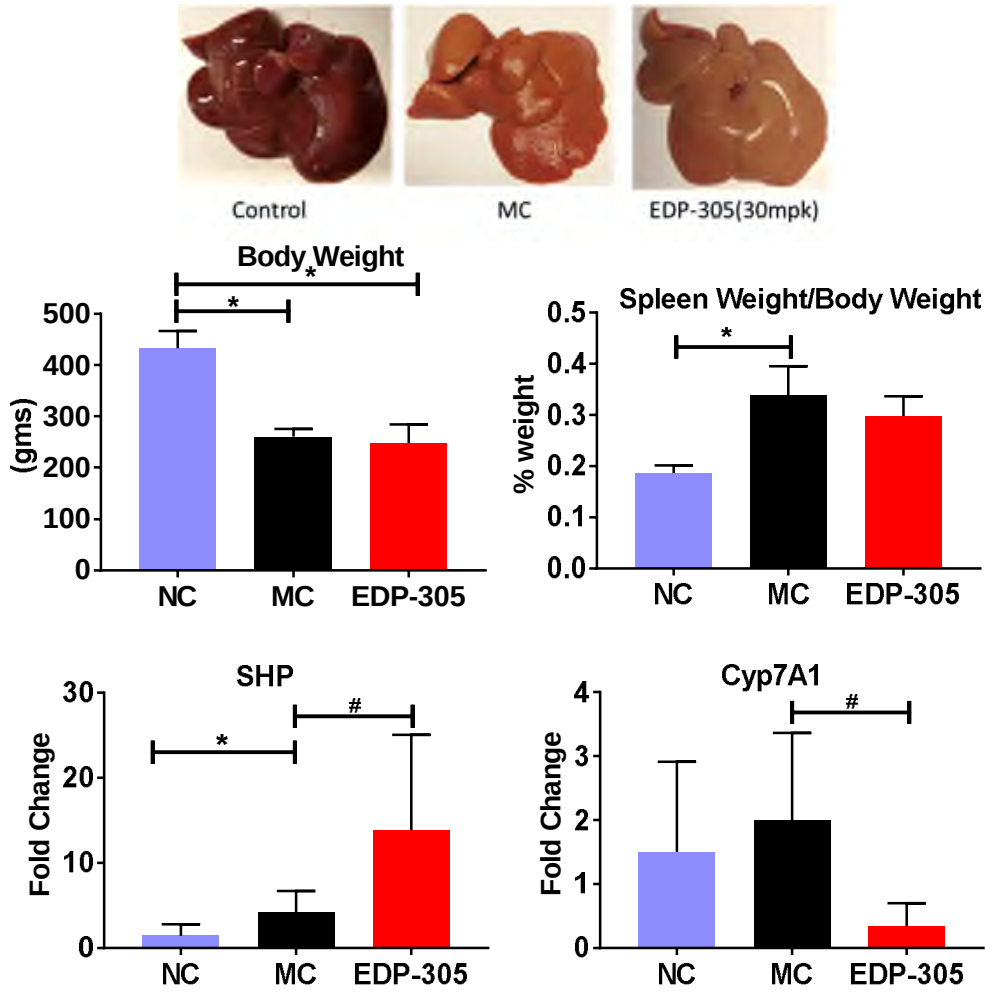
Control
MC
EDP-305(30mpk)
Body Weight
(gms)
Spleen Weight/Body Weight
% weight
SHP
Fold Change
Cyp7A1
Fold Change
NC
MC
EDP-305

EDP-305 was well tolerated in CDAHFD rats as indicated by no change in body weight compared to MC. EDP-305 increased liver *Shp* gene expression and decreased liver *Cyp7a1* gene expression indicative of FXR agonism.

EDP-305 Treatment Improved Liver Function in CDAHFD Rat Model

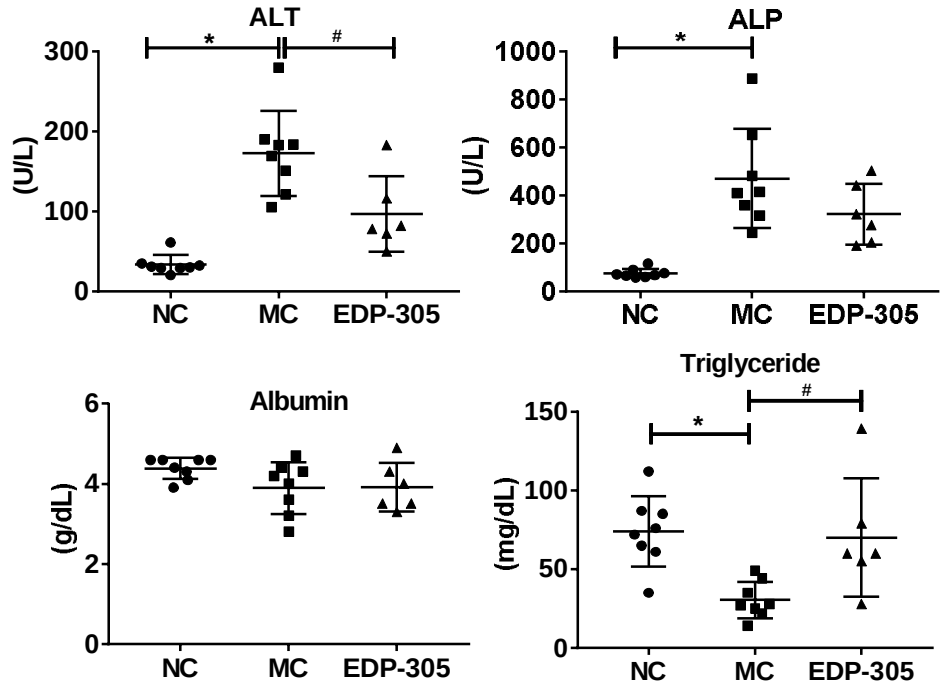
ALT
(U/L)
ALP
(U/L)
Albumin
(g/dL)
Triglyceride
(mg/dL)
NC
MC
EDP-305

EDP-305 treatment decreased serum ALT suggestive of reduced hepatic injury.

EDP-305 Treatment Reduced Fibrosis

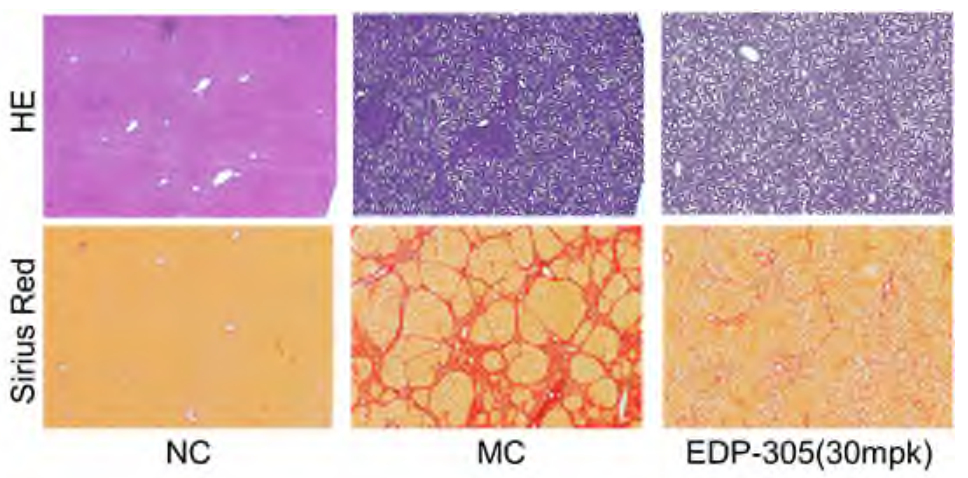
HE
Sirius Red
NC
MC
EDP-305(30mpk)

Rats fed CDAHFD developed nodular cirrhosis. EDP-305 significantly decreased fibrosis in CDAHFD fed rats as assessed by Sirius red staining.

EDP-305 Treatment Reduced Fibrosis

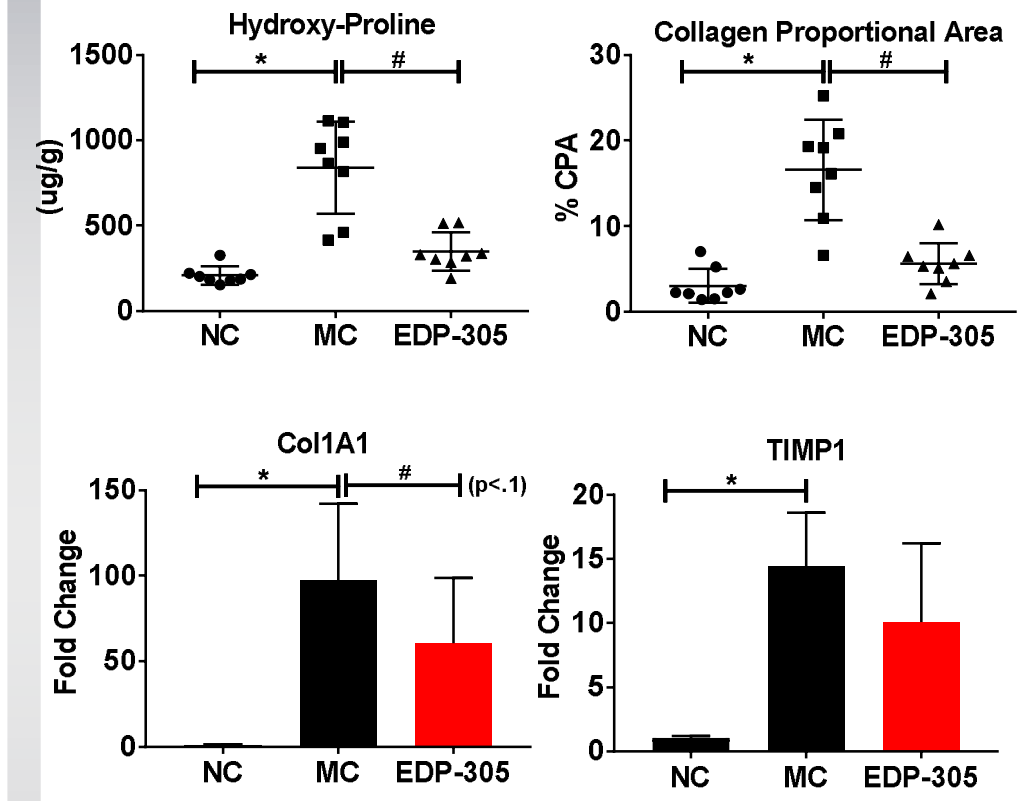
Hydroxy-Proline
(ug/g)
Collagen Proportional Area
% CPA
Col1A1
(p<.1)
Fold Change
TIMP1
Fold Change
NC
MC
EDP-305

EDP-305 30 mpk reduced fibrosis as assessed by morphometric quantification of the collagen proportional area (CPA) in Sirius Red stained liver sections and HPLC measurement of Hydroxy-Proline levels in liver tissue. qPCR further confirmed reduced expression of *Col1A1* after EDP-305 treatment.

Conclusion

In a rat model of NASH cirrhosis that more closely resembles the human histology, EDP-305 reduced fibrosis progression.

These results suggest that EDP-305 may have potent anti-fibrotic effects in late-stage F3/4 patients.